Humoral immune response against SARS-CoV-2 variants (Omicron BA.1, Beta, Delta and XBB.1.5) of PHH-1V booster vaccine in subjects previously vaccinated with a mRNA vaccine. Results of a randomised controlled trial up to 6 and 12 months.

A. Moros[1], A. Barreiro[1],H. Bernard[2], R. Abu Taleb[2] T. Prenafeta[1], L. Madrenas[1], M. Cañete[1], J. Corominas[1], T. Prat[1], L. Ferrer[1]

[1]HIPRA, Girona. [2]VERISTAT, Barcelona.

Background

There is still a need for broad-spectrum vaccines that may cross protect from new genetic variants of SARS-CoV-2 and provide a long-lasting immune response. PHH-1V (Bimervax®, HIPRA) is a bivalent recombinant protein vaccine based on a heterodimer consisting of the receptor binding domain of two SARS-CoV-2 variants, Beta and Alpha, designed to boost immunogenicity against SARS-CoV-2 in fully vaccinated adult individuals.

Objective

To assess immunogenicity of a booster vaccination with PHH-1V (Bimervax®, HIPRA) compared with BNT162b2 mRNA vaccine (Comirnaty®, Pfizer/BioNTech) in adults fully vaccinated with primary series against COVID-19.

Methods

HIPRA-HH-2 study (NCT05142553) is a Phase IIb, double-blind, randomised, active-controlled, multi-centre clinical trial. 765 participants with a complete prime course against COVID-19 with BNT162b2 were randomised (2:1) to receive a booster dose of PHH-1V or BNT162b2.

Population

Randomisation (2:1)

N=765

PHH-1V group
N=513 (N=101 for XBB.1.5)

BNT162b2 group
N=252 (N=51 for XBB.1.5)

≥18 years old (7.3 % ≥65 years old)
Prime vaccination: 2 doses of BNT162b2 (at least 182 days ago)
Naïve subjects at the time of vaccination
White/Caucasian, Hispanic, Asian population included

Procedure

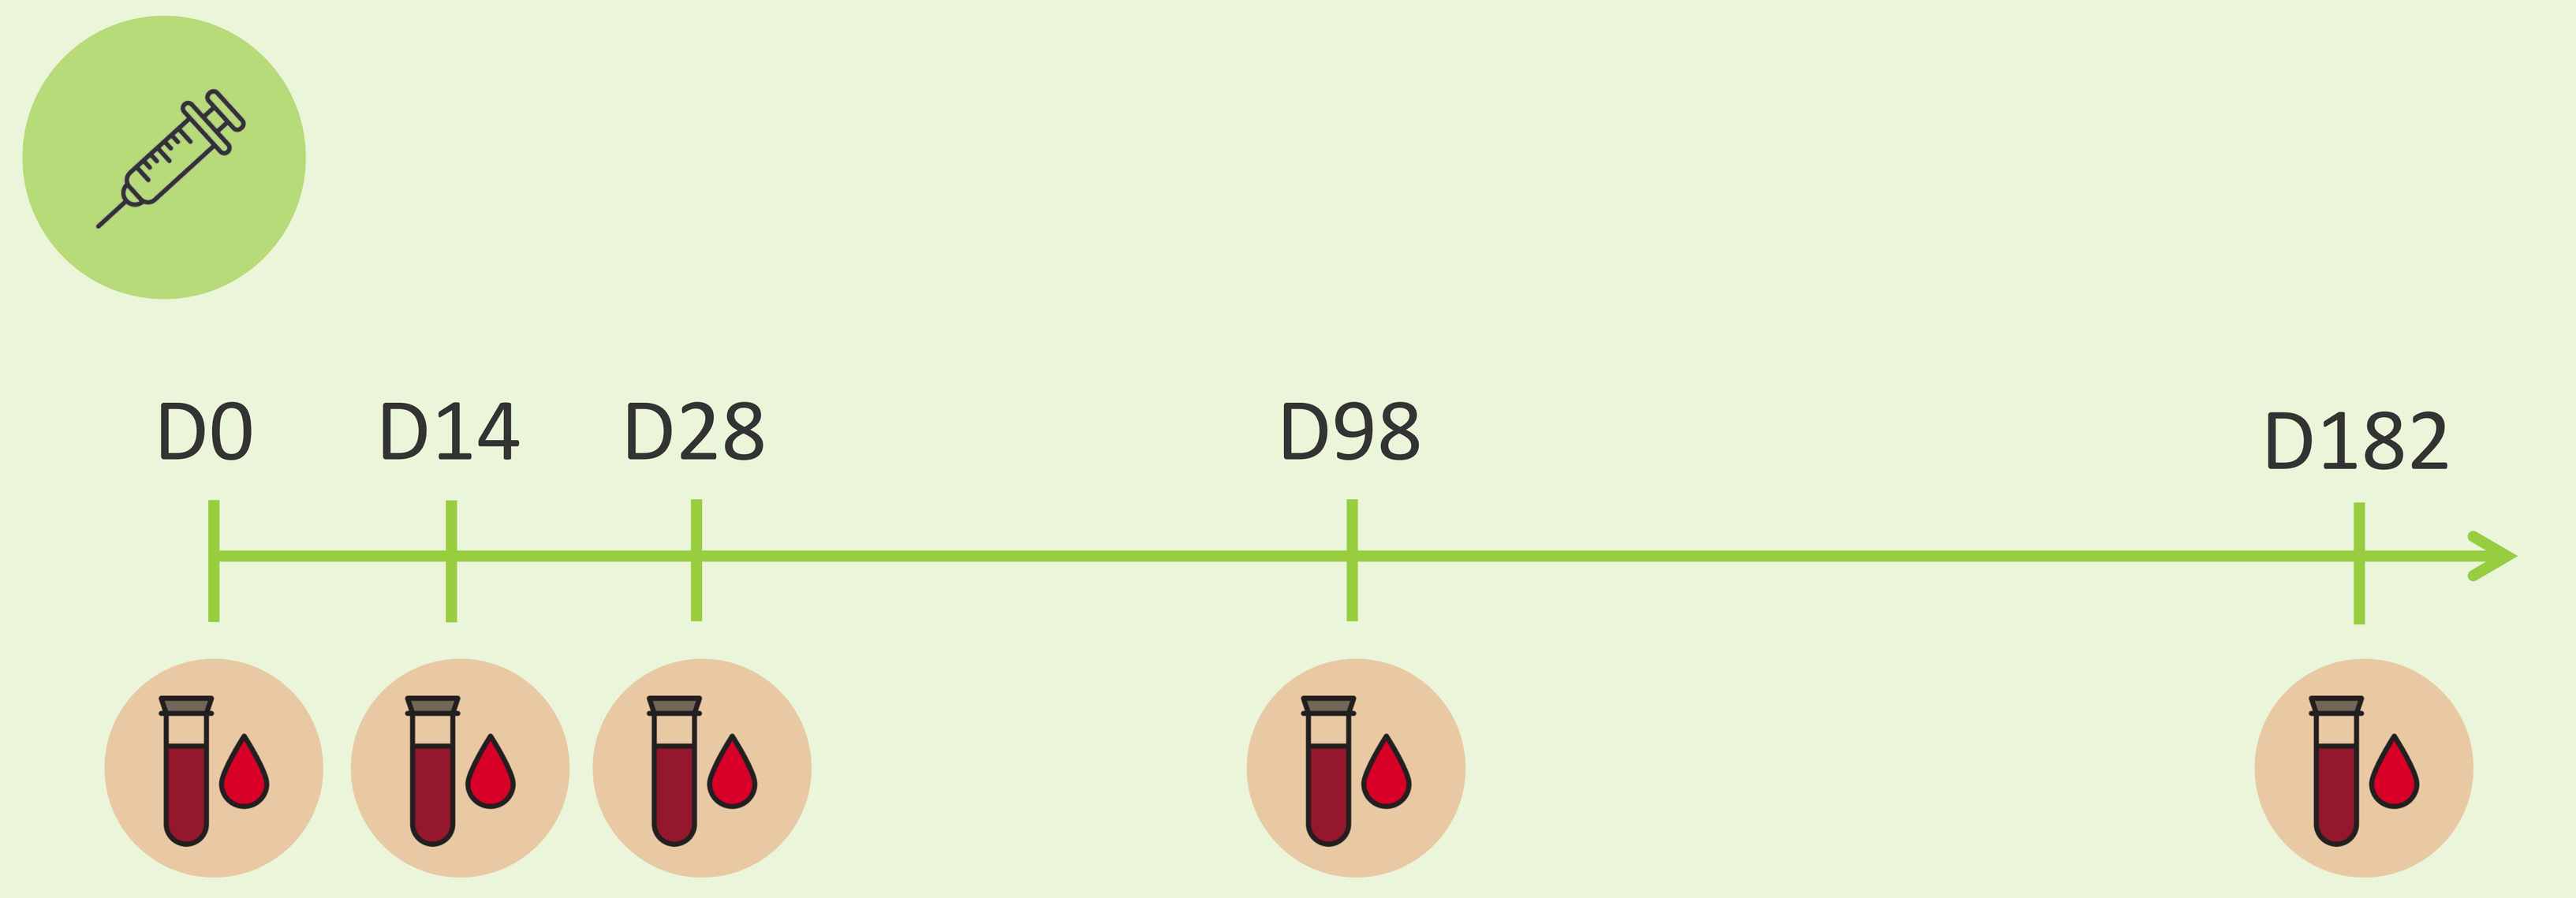
D0
D14
D28
D98
D182

Humoral immunogenicity was measured by changes in levels of neutralising antibodies (PBNA) after PHH-1V (N=513) or BNT162b2 (N=2452) boost at Day 14, 28, 98, 182 against Wuhan-Hu-1, Beta, Delta and Omicron BA.1 variants. The same analysis after PHH-1V (N=101) or BNT162b2 (N=52) boost was performed against Omicron XBB.1.5 at Day 14 , 182 and 365. The number of subjects with SARS-CoV-2 infection after the booster was also assessed up to 12 months.

Results

PHH-1V Booster is superior to BNT162b2 at 6 months

Both PHH-1V and BNT162b2 vaccines elicited a strong neutralising antibody response 14 days after the booster against all variants tested. PHH-1V induced a sustained neutralising antibody response up to 6 months against all variants. When compared to BNT162b2, superiority of PHH-1V was demonstrated against Wuhan, Beta, Delta and Omicron BA.1 variants 6 months after the booster (Day 182) (GMT ratios: 0.62 (*p*<0.0001), 0.70 (*p*<0.0001), 0.55 (*p*<0.0001) and 0.76 (*p*=0.0028), respectively) (Figure 1).

PHH-1V Booster induces immune response to XBB.1.5

Results show that both BNT162b2 and PHH-1V induce a strong and sustained neutralizing antibodies response against Omicron XBB.1.5, PHH-1V response being numerically higher compared to BNT162b (GMF ratios for PHH-1V and BNT162b2 respectively are: 7.58 and 7.17 at D14; 6 and 5.42 at D182; 5.1 and 4.82 at D365) (Figure 2 and Table 1). Compared to the humoral response induced by PHH-1V vaccination against previous variants[1], the XBB.1.5 neutralizing antibody levels are lower, which is in line with the results obtained with boosters with other vaccines against the XBB.1.5 subvariant[2,3,4].

Neutralizing antibodies titres against different SARS-CoV-2 variants

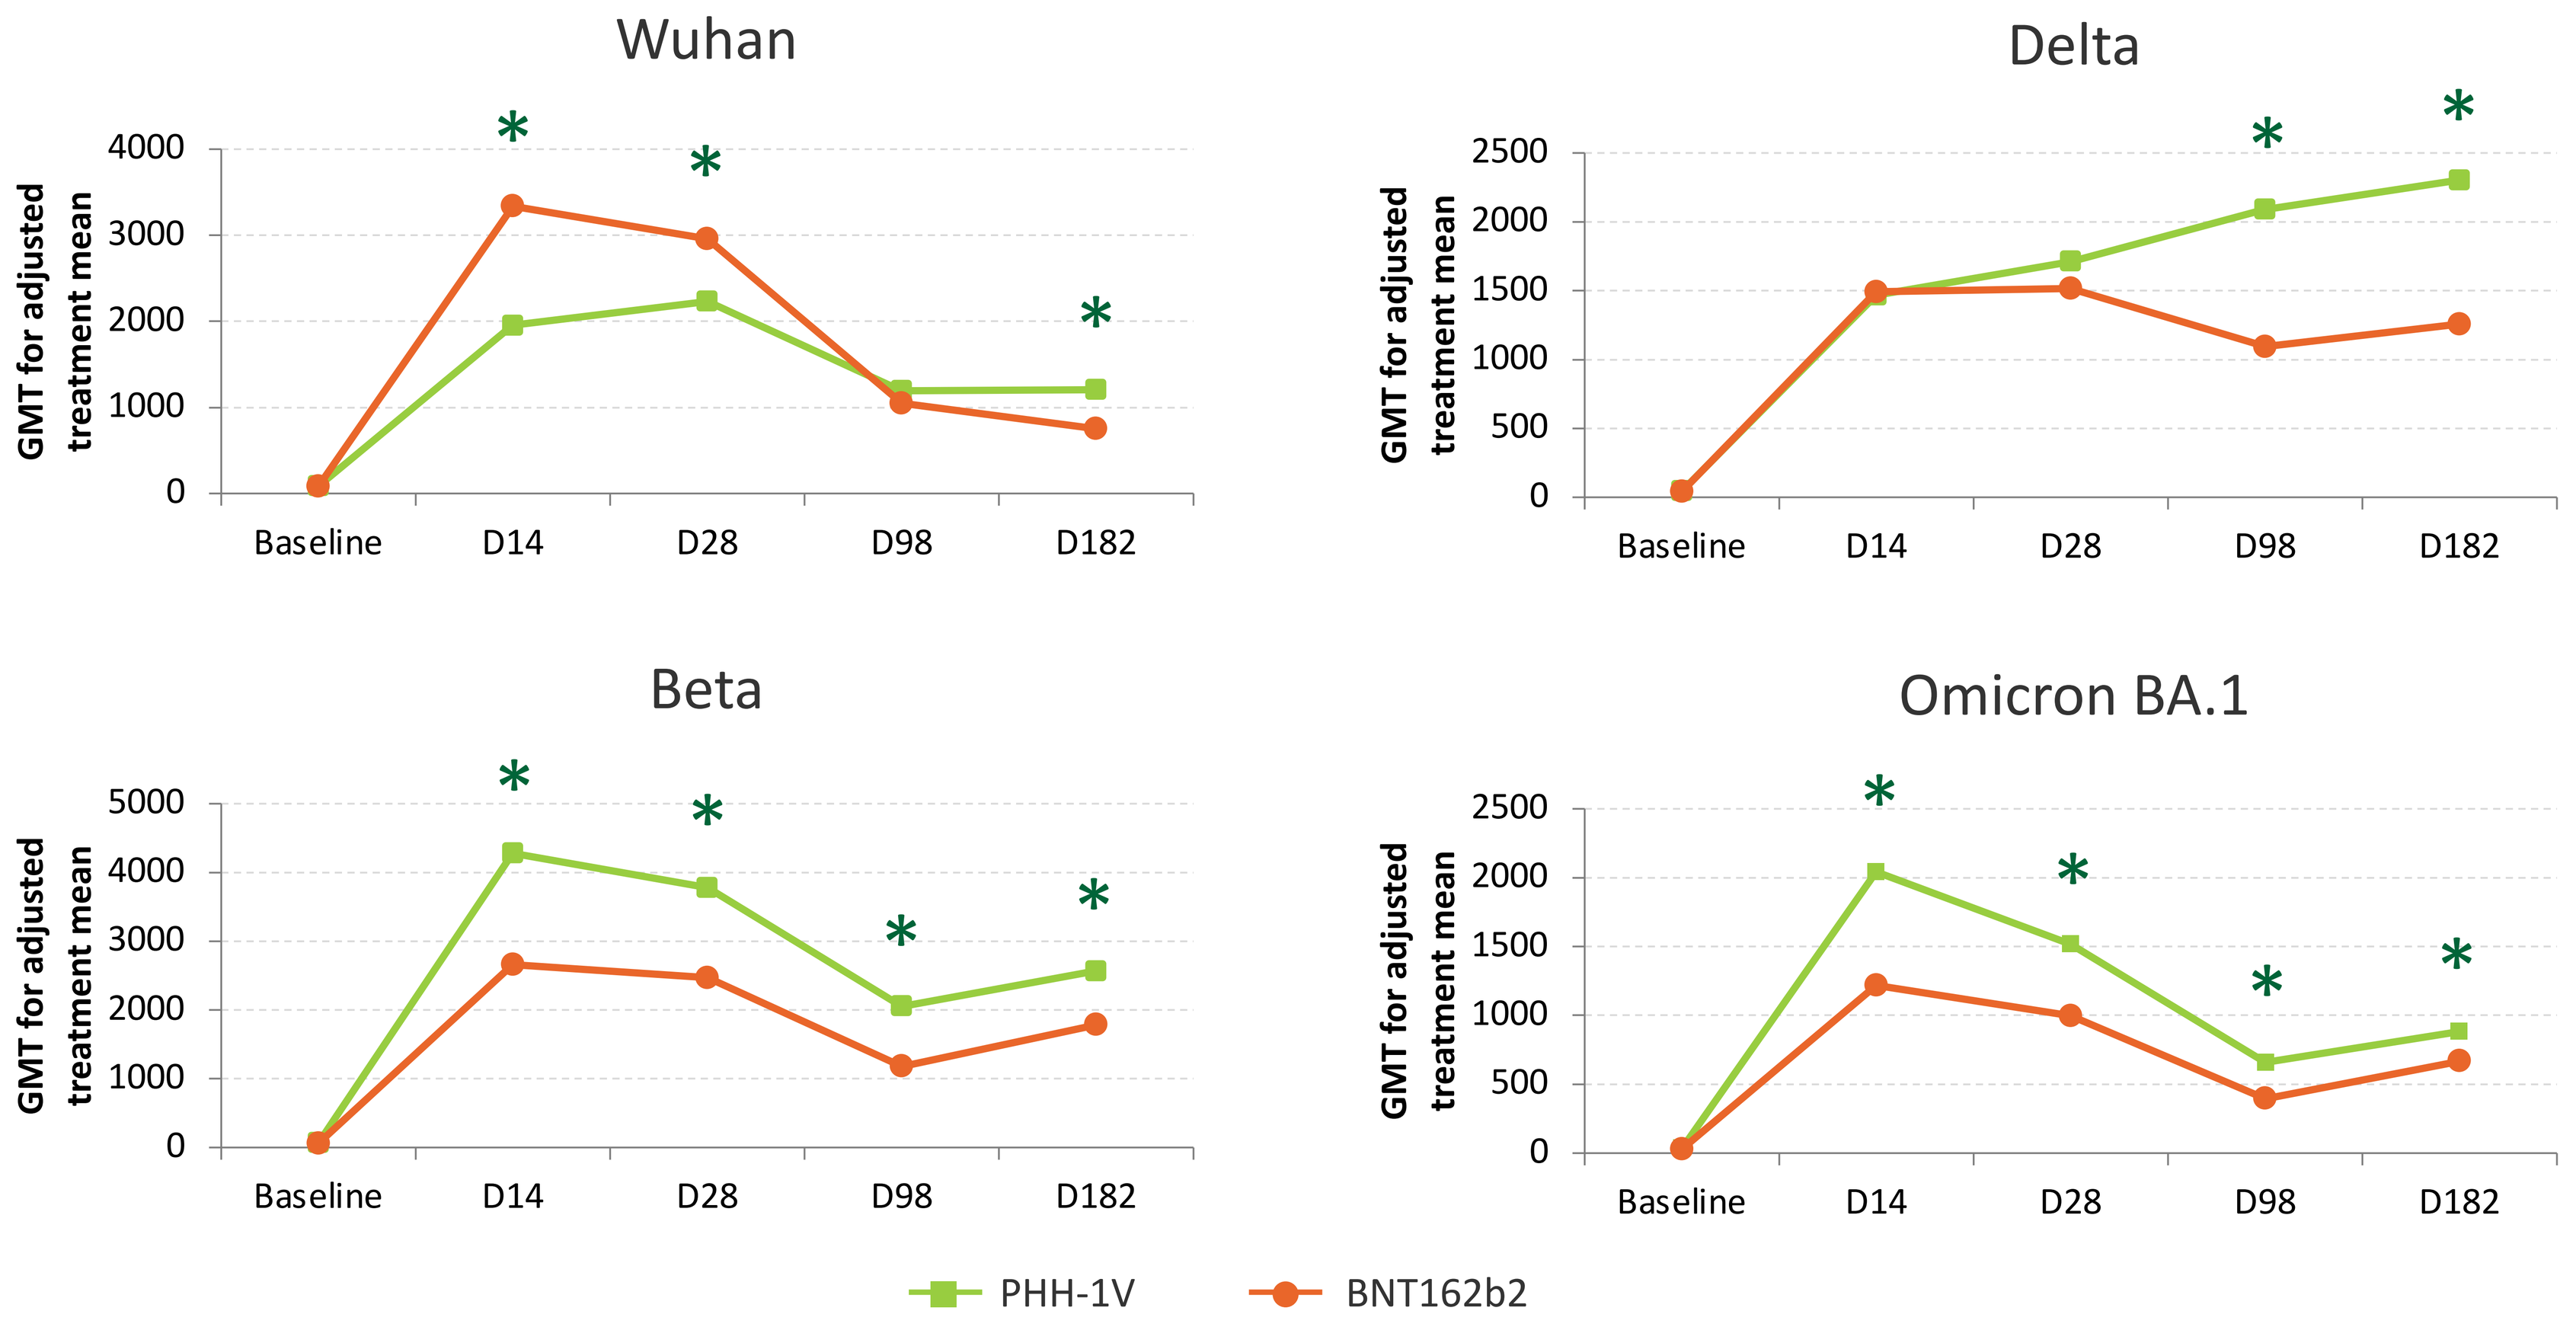
Wuhan
Delta
Beta
Omicron BA.1
GMT for adjusted treatment mean
Baseline
D14
D28
D98
D182
PHH-1V
BNT162b2

Figure 1. Neutralising antibody titres (GMT for adjusted treatment mean) of PHH-1V and BNT162b2 vaccines against different SARS-CoV-2 variants over time.
Statistically significant differences are shown as * for p<0.05.
Abbreviations: GMT = Geometric Mean Titre.

Neutralizing antibodies titres against Omicron XBB.1.5

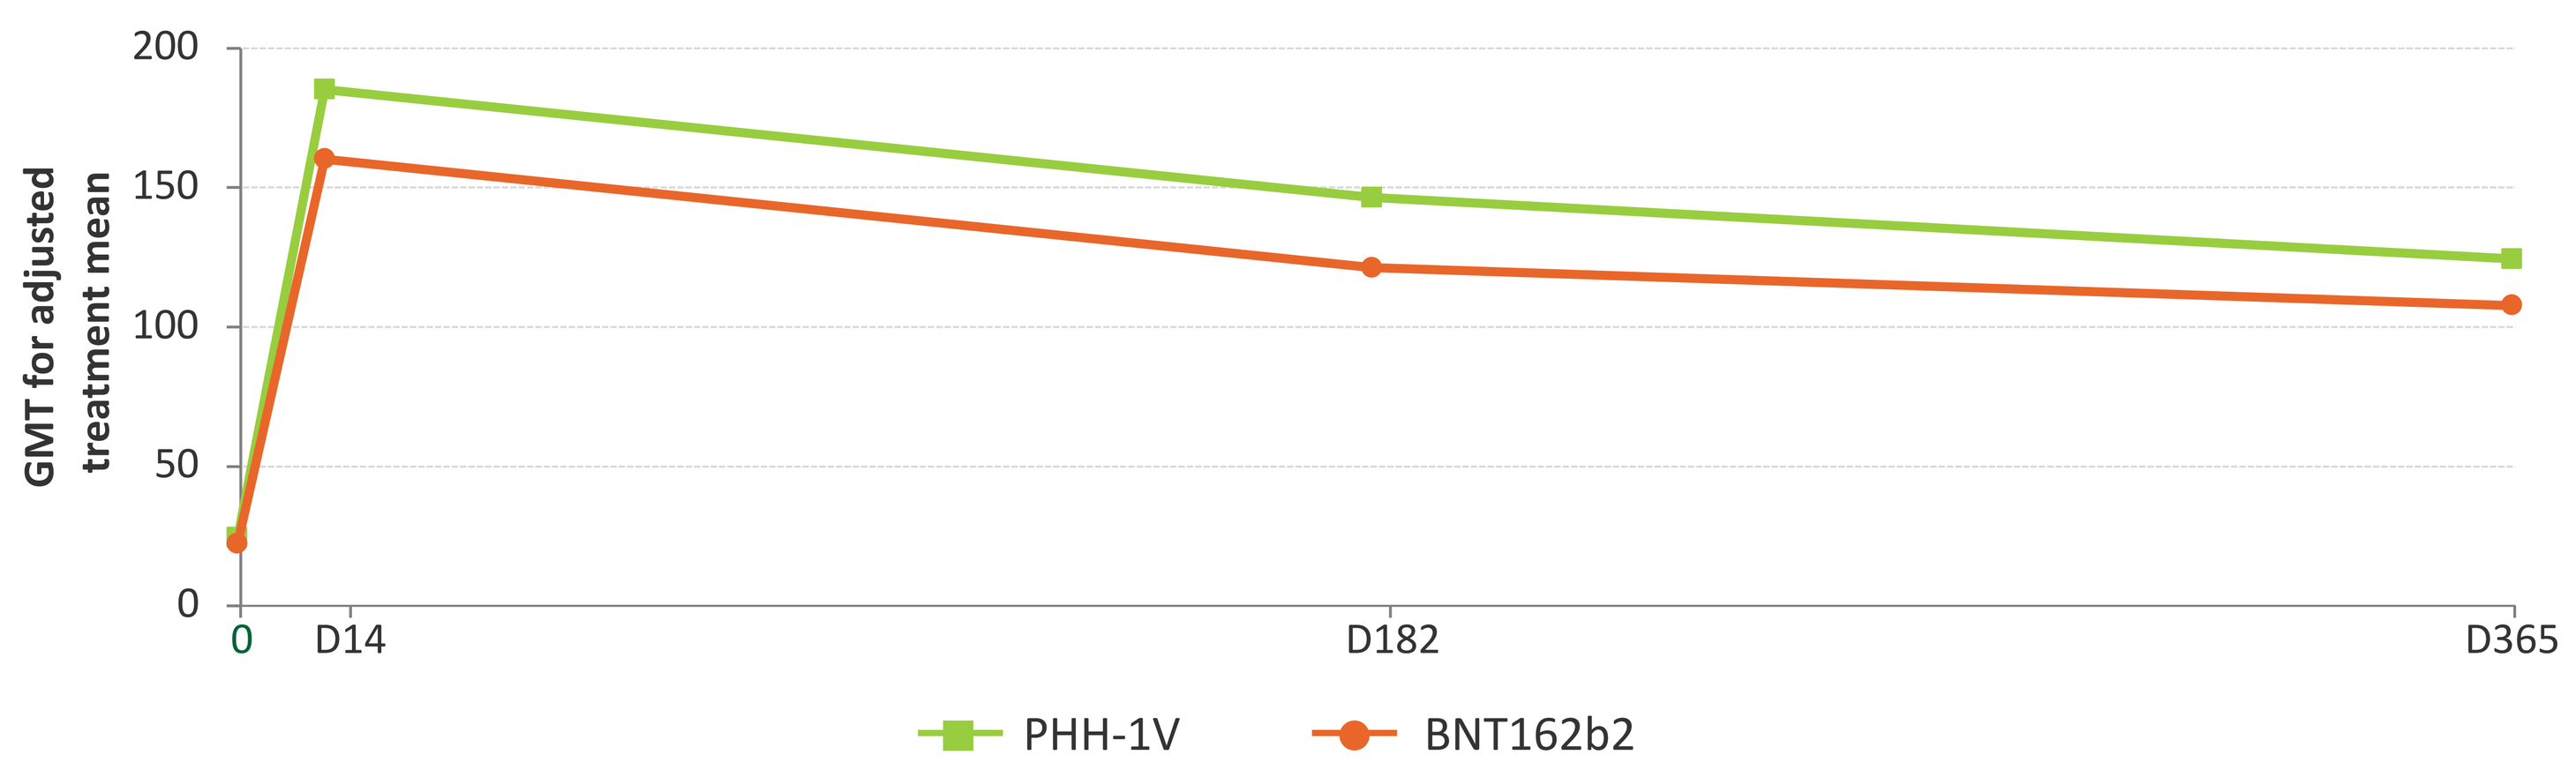
GMT for adjusted treatment mean
0
D14
D182
D365
PHH-1V
BNT162b2

Figure 2. Neutralising antibody titres (GMT for adjusted treatment mean) of PHH-1V and BNT162b2 vaccines against SARS-CoV-2 Omicron XBB.1.5 over time. Statistical analysis is in progress.
Abbreviations: GMT = Geometric Mean Titre. PBNA= Pseudovirus Antiboidies assay.

	Omicron XBB.1.5						
	Baseline	D14		D182		D365	
	GMT	GMT	GMFR	GMT	GMFR	GMT	GMFR
3rth dose PHH-1V (N=101)	24.42	185.21	7.58	146.50	6	124.46	5.1
3rth dose BNT162b (N=51)	22.35	160.23	7.17	121.24	5.42	107.75	4.82

Table 1. Neutralising antibody titres (GMT for adjusted treatment mean) of PHH-1V and BNT162b2 vaccines against SARS-CoV-2 Omicron XBB.1.5 over time.
Abbreviations: GMT = Geometric Mean Titre, GMFR= Geometric Mean Fold Rise calculated from baseline.

During all the study period (12-month follow-up), only non-severe COVID-19 cases were reported (65.1 % and 58.5 % with PHH-1V and BNT162b2, respectively). PHH-1V demonstrates a very good safety profile overall.

Conclusions

PHH-1V (Bimervax®, HIPRA) as a booster elicits a strong and sustained neutralising antibody response against all tested variants in individuals with a complete primary course of mRNA vaccine and protects from severe disease.

Although the increase in antibody titres against XBB.1.5 is lower than those against previous SARS-CoV-2 variants (D614G, Beta, Delta, Omicron BA.1 and BA.4/5)[1], the response generated by the BIMERVAX® booster appears to be sufficient to protect against severe disease.

References

1. Corominas J, Garriga C, Prenafeta A, Moros A, Cañete M, Barreiro A, et al. Safety and immunogenicity of the protein-based PHH-1V compared to BNT162b2 as a heterologous SARS-CoV-2 booster vaccine in adults vaccinated against COVID-19: a multicentre, randomised, double-blind, non-inferiority phase IIb trial. Lancet Reg Health Eur. 2023; 28:100613. doi: https://doi.org/10.1016/j.lanepe.2023.100613
2. Sutandhio S, Furukasa K, Kurahashi Y, Marini MI, Effendi GB, Hasegawa N, et al. Fourth mRNA vaccination increases cross-neutralizing antibody titers against SARS-CoV-2 variants, including BQ.1.1 and XBB, in a very elderly population. J Infect Public Health. 2023;16(7):1064-1072. doi: https://doi.org/10.1016/j.jiph.2023.05.004.
3. Kurhade C, Zou J, Xia H, Liu M, Chang HC, Ren P, et al. Low neutralization of SARS-CoV-2 Omicron BA.2.75.2, BQ.1.1 and XBB.1 by parental mRNA vaccine or a BA.5 bivalent booster. Nat Med. 2023;29(2):344-347. doi: https://doi.org/10.1038/s41591-022-02162-x.
4. Devasundaram S, Terpos E, Rosati M, Ntanasis-Stathopoulos I, Bear J, Burns R, et al. XBB.1.5 neutralizing antibodies upon bivalent COVID-19 vaccination are similar to XBB but lower than BQ.1.1. Am J Hematol. 2023 May;98(5):E123-E126. doi: https://doi.org/10.1002/ajh.26887